Imetelstat achieved prolonged, continuous transfusion independence in patients with heavily transfused non-del(5q) lower-risk myelodysplastic syndromes relapsed/refractory to erythropoiesis stimulating agents within the IMerge phase 2 trial

U. Platzbecker,[1] R. Komrokji,[2] P. Fenaux,[3] M.A. Sekeres,[4] M.R. Savona,[5] Y.F. Madanat,[6] K. Van Eygen,[7] A. Raza,[8] U. Germing,[9] S. Shah,[10] L. Sun,[10] A. Ikin,[10] F. Feller,[10] A. Zeidan,[11]* V. Santini[12]*

[1]University Clinic Leipzig, Leipzig, Germany; [2]Moffitt Cancer Center, Tampa, FL, USA; [3]Hôpital Saint-Louis, Université Paris Diderot, Paris, France; [4]Sylvester Cancer Center, University of Miami, Miami, FL, USA; [5]Vanderbilt University Medical Center, Nashville, TN, USA; [6]University of Texas Southwestern Medical Center, Dallas TX, USA; [7]Algemeen Ziekenhuis Groeninge, Kortrijk, Belgium; [8]Columbia University Medical Center, New York, NY, USA; [9]Klinik für Hämatologie, Onkologie, and Klinischelmmunologie, Universitätsklinik Düsseldorf, Heinrich-Heine-Universität, Düsseldorf, Germany; [10]Geron Corporation, Parsippany, NJ, USA; [11]Yale School of Medicine, New Haven, CT, USA; [12]MDS Unit, AOU Careggi-University of Florence, Florence, Italy. *Contributed equally

Introduction

- Novel efficacious and safe treatments are needed for patients with lower-risk myelodysplastic syndromes (LR-MDS) who are red blood cell (RBC) transfusion dependent (TD), and have relapsed after or are refractory (R/R) to erythropoiesis-stimulating agents (ESA)

Fig. 1. Imetelstat Binds to RNA Template Preventing Maintenance of Telomeres

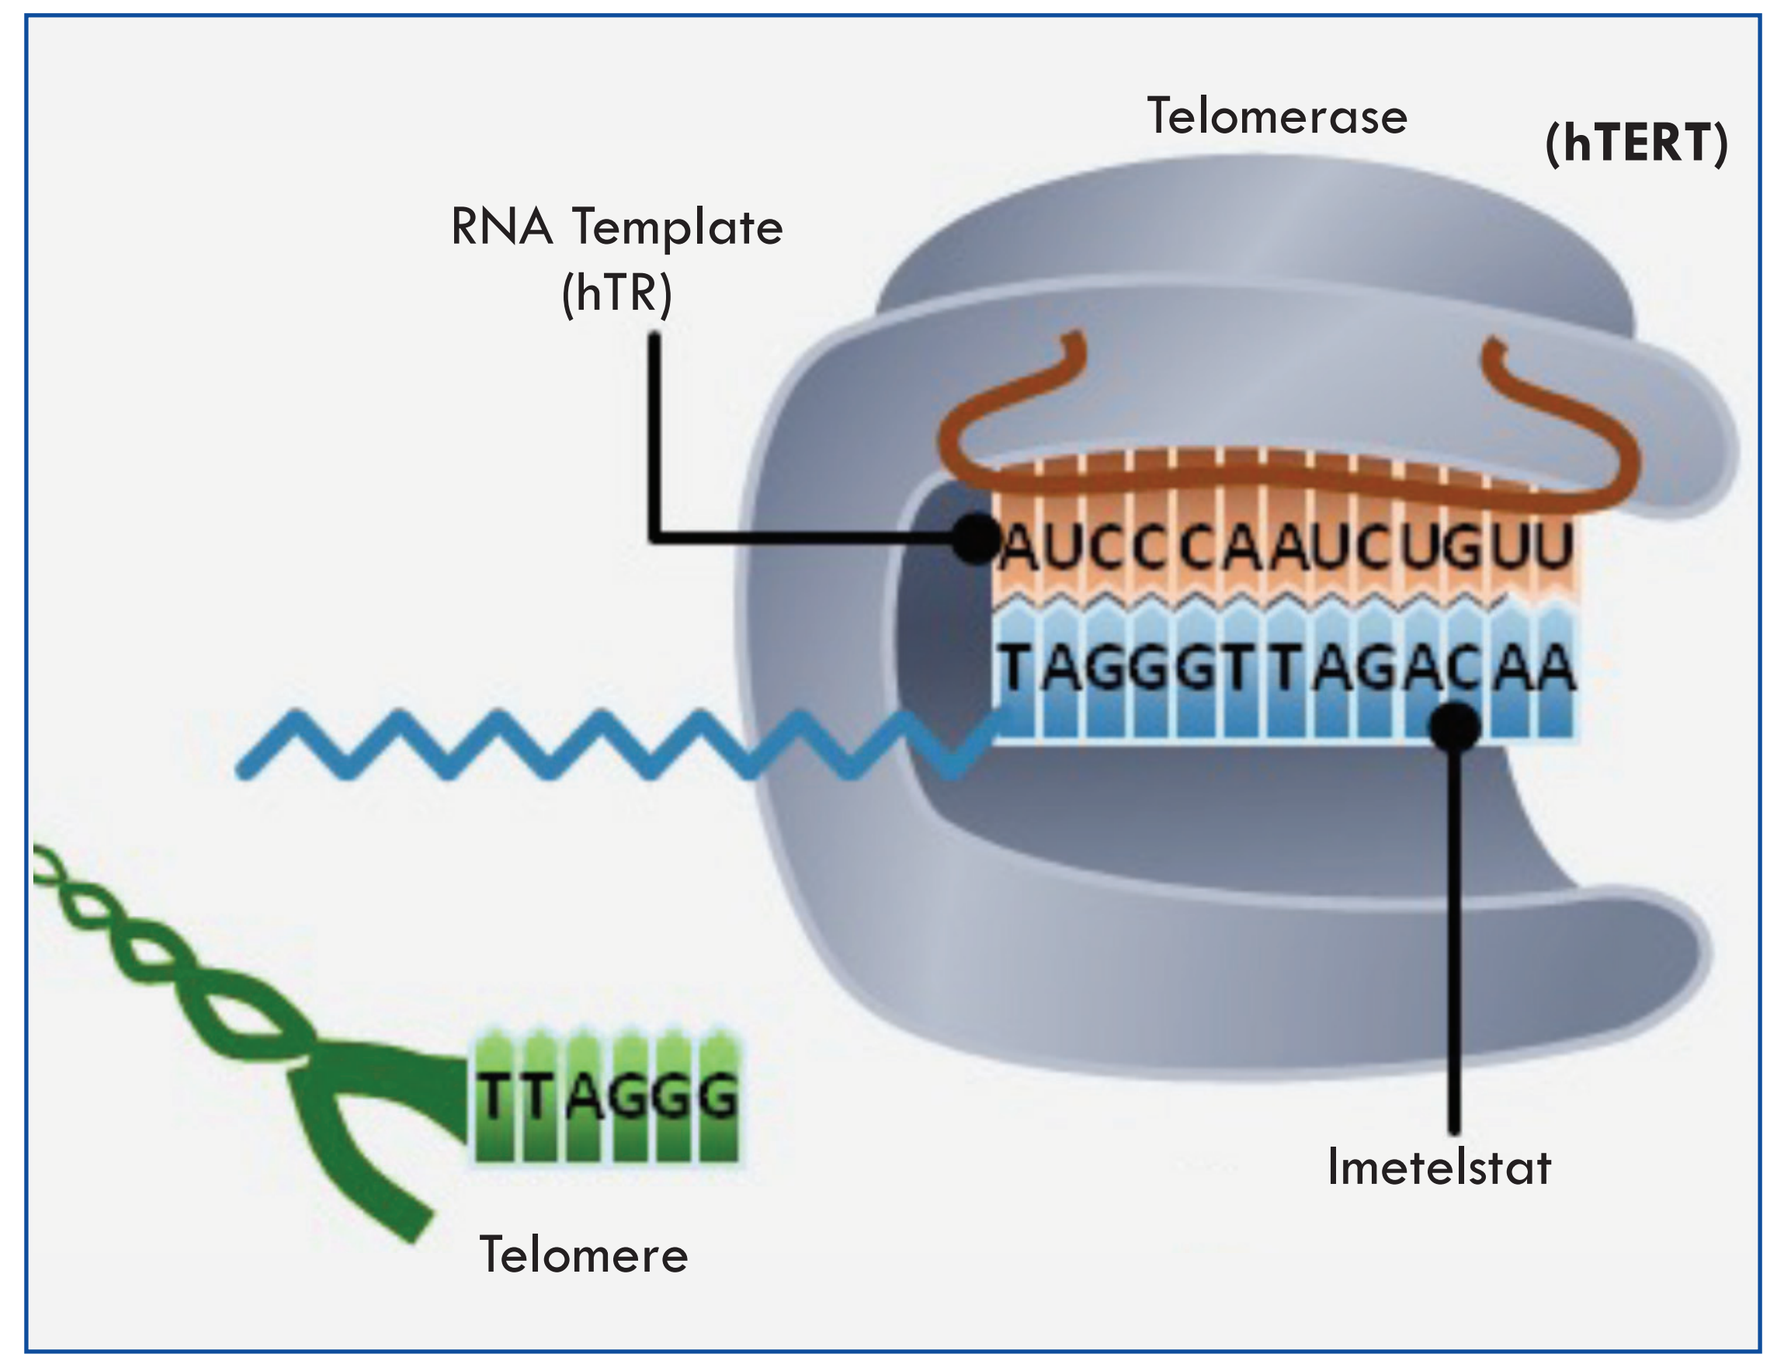
Telomerase
(hTERT)
RNA Template
(hTR)
AUCCCAAUCUGUU
TAGGGTTAGACAA
TTAGGG
Telomere
Imetelstat

hTERT, human telomerase reverse transcriptase; hTR, catalytic component.

- MDS are a heterogeneous group of myeloid cancers characterized by clonal proliferation of malignant stem and progenitor cells, peripheral-blood cytopenias, myelodysplasia, and an increased risk of progression to acute myeloid leukemia (AML)
- Imetelstat, a first-in-class telomerase inhibitor, targets cells with high telomerase activity and human telomerase reverse transcriptase (hTERT) expression, both of which have been reported in MDS
- Being a direct and competitive inhibitor of telomerase activity[1,2] (**Fig. 1**), imetelstat has disease-modifying potential to selectively kill malignant stem and progenitor cells, enabling recovery of blood cell production[3,4] (**Fig. 2**)

Fig. 2. Imetelstat Selectively Kills Malignant Stem and Progenitor Cells Enabling Recovery of Blood Cell Production

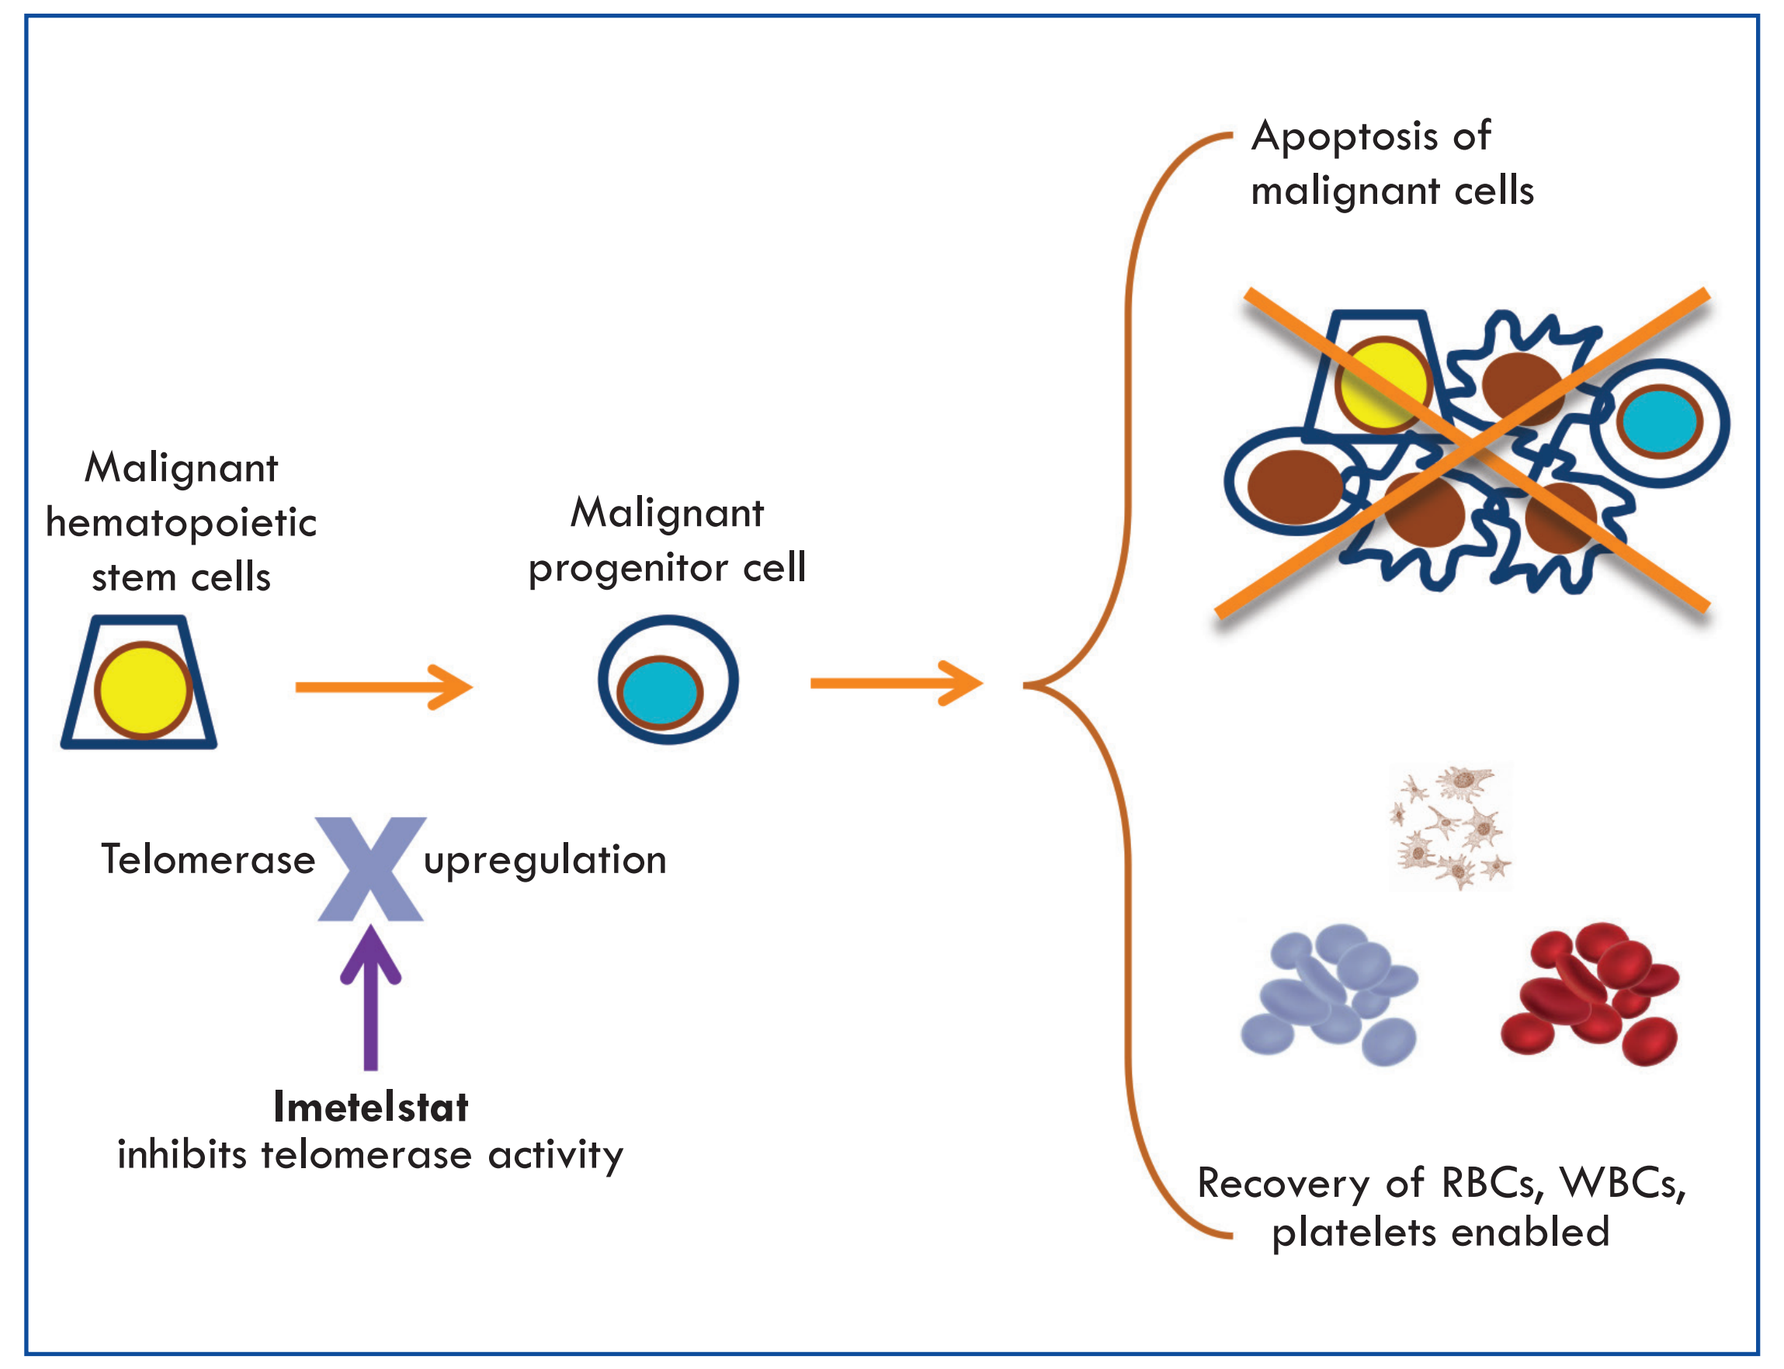
Apoptosis of
malignant cells
Malignant
hematopoietic
stem cells
Malignant
progenitor cell
Telomerase
upregulation
Imetelstat
inhibits telomerase activity
Recovery of RBCs, WBCs,
platelets enabled

RBC, red blood cell; WBC, white blood cell.

- The IMerge (NCT02598661) phase 2 trial previously reported meaningful and durable transfusion independence (TI) with imetelstat treatment[5,6]
 - Of the 57 patients treated, 38 patients were non-del(5q) and lenalidomide (len)/ hypomethylating agent (HMA) naive (target patient population)
 - A longer TI duration was seen in this target population (median, 88 weeks) vs all 57 treated patients (median, 65 weeks)
 - An 8-week and 24-week TI response was reached by 16 patients (42%) and 12 patients (32%), respectively
 - After 57 months of follow-up, 11 patients (29%) from the target population (N=38) had continuous TI for ≥1 year while on imetelstat treatment[6]
 - These patients represented 69% of the ≥8-week TI responders, 92% of the ≥24-week TI responders, and 37% (10/27) of the patients with MDS ring sideroblast-positive (RS+) treated

Objective

- To determine the characteristics and clinical benefits of the 11 patients within the target patient population who, after 57 months of follow-up, had continuous TI for ≥1 year while on imetelstat

Trial Status

- This IMerge (MDS3001) trial is registered at ClinicalTrials.gov (NCT02598661)
- Enrollment is complete for IMerge phase 3
- Top line results were disclosed to investors January 4, 2023, and will be presented at future congresses[7]
- For further information please visit https://www.geron.com/patients/imerge-study or contact: mds3001-info@geron.com

Results

- The cutoff date for all analyses was October 13, 2022
- All 11 patients had previously been treated with ESAs; additional baseline characteristics are also provided (**Table 1**)
- The median treatment duration for the 11 patients was 126.1 weeks (range, 70.1–168.1) with a median of 27 cycles (range, 18–40), and a median relative dose intensity[a] of 98.9% (range, 85.5–102.4)
 - The median time on trial[b] was 57.3 months (range, 19.0–57.8)
- The median prior RBC transfusion burden for the 11 patients was 6 (range, 4-14) units over 8 weeks (**Table 1**)

Efficacy

- The median onset of 8-week TI was 9.29 weeks (range, 3.3–40.7) (**Fig. 3**) and median TI duration was 92.4 weeks (95% CI, 69.57–140.86) (**Fig. 4A**) with increased Hgb levels (**Fig. 4B**)
- Median PFS for the TI ≥1-year responders (n=11) was 34.2 months (95% CI, 25.10–39.16) (**Fig. 5A**)
- Median OS for the TI ≥1-year responders was 56.1 months (95% CI, 29.4–not evaluable) (**Fig. 5B**)
- PFS and OS of the ≥1-year TI responders and target population are summarized in **Table 2**
- After a median follow-up of 57 months, no progressions to AML were observed among the ≥1-year TI responders

[a]Defined as the total actual dose/total planned dose. [b]Defined as the interval between trial day 1 and the date of death (censored) or last day on the trial; based on the Kaplan-Meier method.

Table 1. Baseline Characteristics of Patients With TI ≥1-Year Treated With Imetelstat Compared With the Overall Target Population

Baseline characteristic	Patients with TI ≥1 year (n=11)	Target population (N=38)
≥2 Years since initial diagnosis, n (%)	10 (90.9)	28 (73.7)
IPSS category, n (%)		
Low	5 (45.5)	24 (63.2)
Intermediate-1 risk	6 (54.5)	14 (36.8)
MDS-RS+, n (%)	10 (90.9)	27 (71.1)
Normal karyotype, n (%)	7 (63.6)	28 (73.7)[a]
Mutations at baseline, n (%)		
SF3B1	11 (100)	27 (71.1)[b]
Other	4 (36.4)	13 (34.2)[b]
Prior ESA, n (%)	11 (100)	34 (89.5)
Prior luspatercept, n (%)	2 (18.2)	6 (15.8)
Median prior RBC transfusion burden (over 8 weeks) prior to trial treatment, units (range)	6.0 (4–14)	8.0 (4–14)

[a]Thirty-four patients had karyotyping results. [b]Baseline mutation samples were collected in 31 patients, 28 out of 31 (90.3%) had a mutation detected. ESA, erythropoiesis-stimulating agent; IPSS, International Prognostic Scoring System; MDS, myelodysplastic syndromes; RBC, red blood cell; RS+, ring sideroblast-positive; TI, transfusion independence.

Fig. 3. Patients With LR-MDS Treated With Imetelstat Achieved Sustained, Continuous TI ≥1 Year

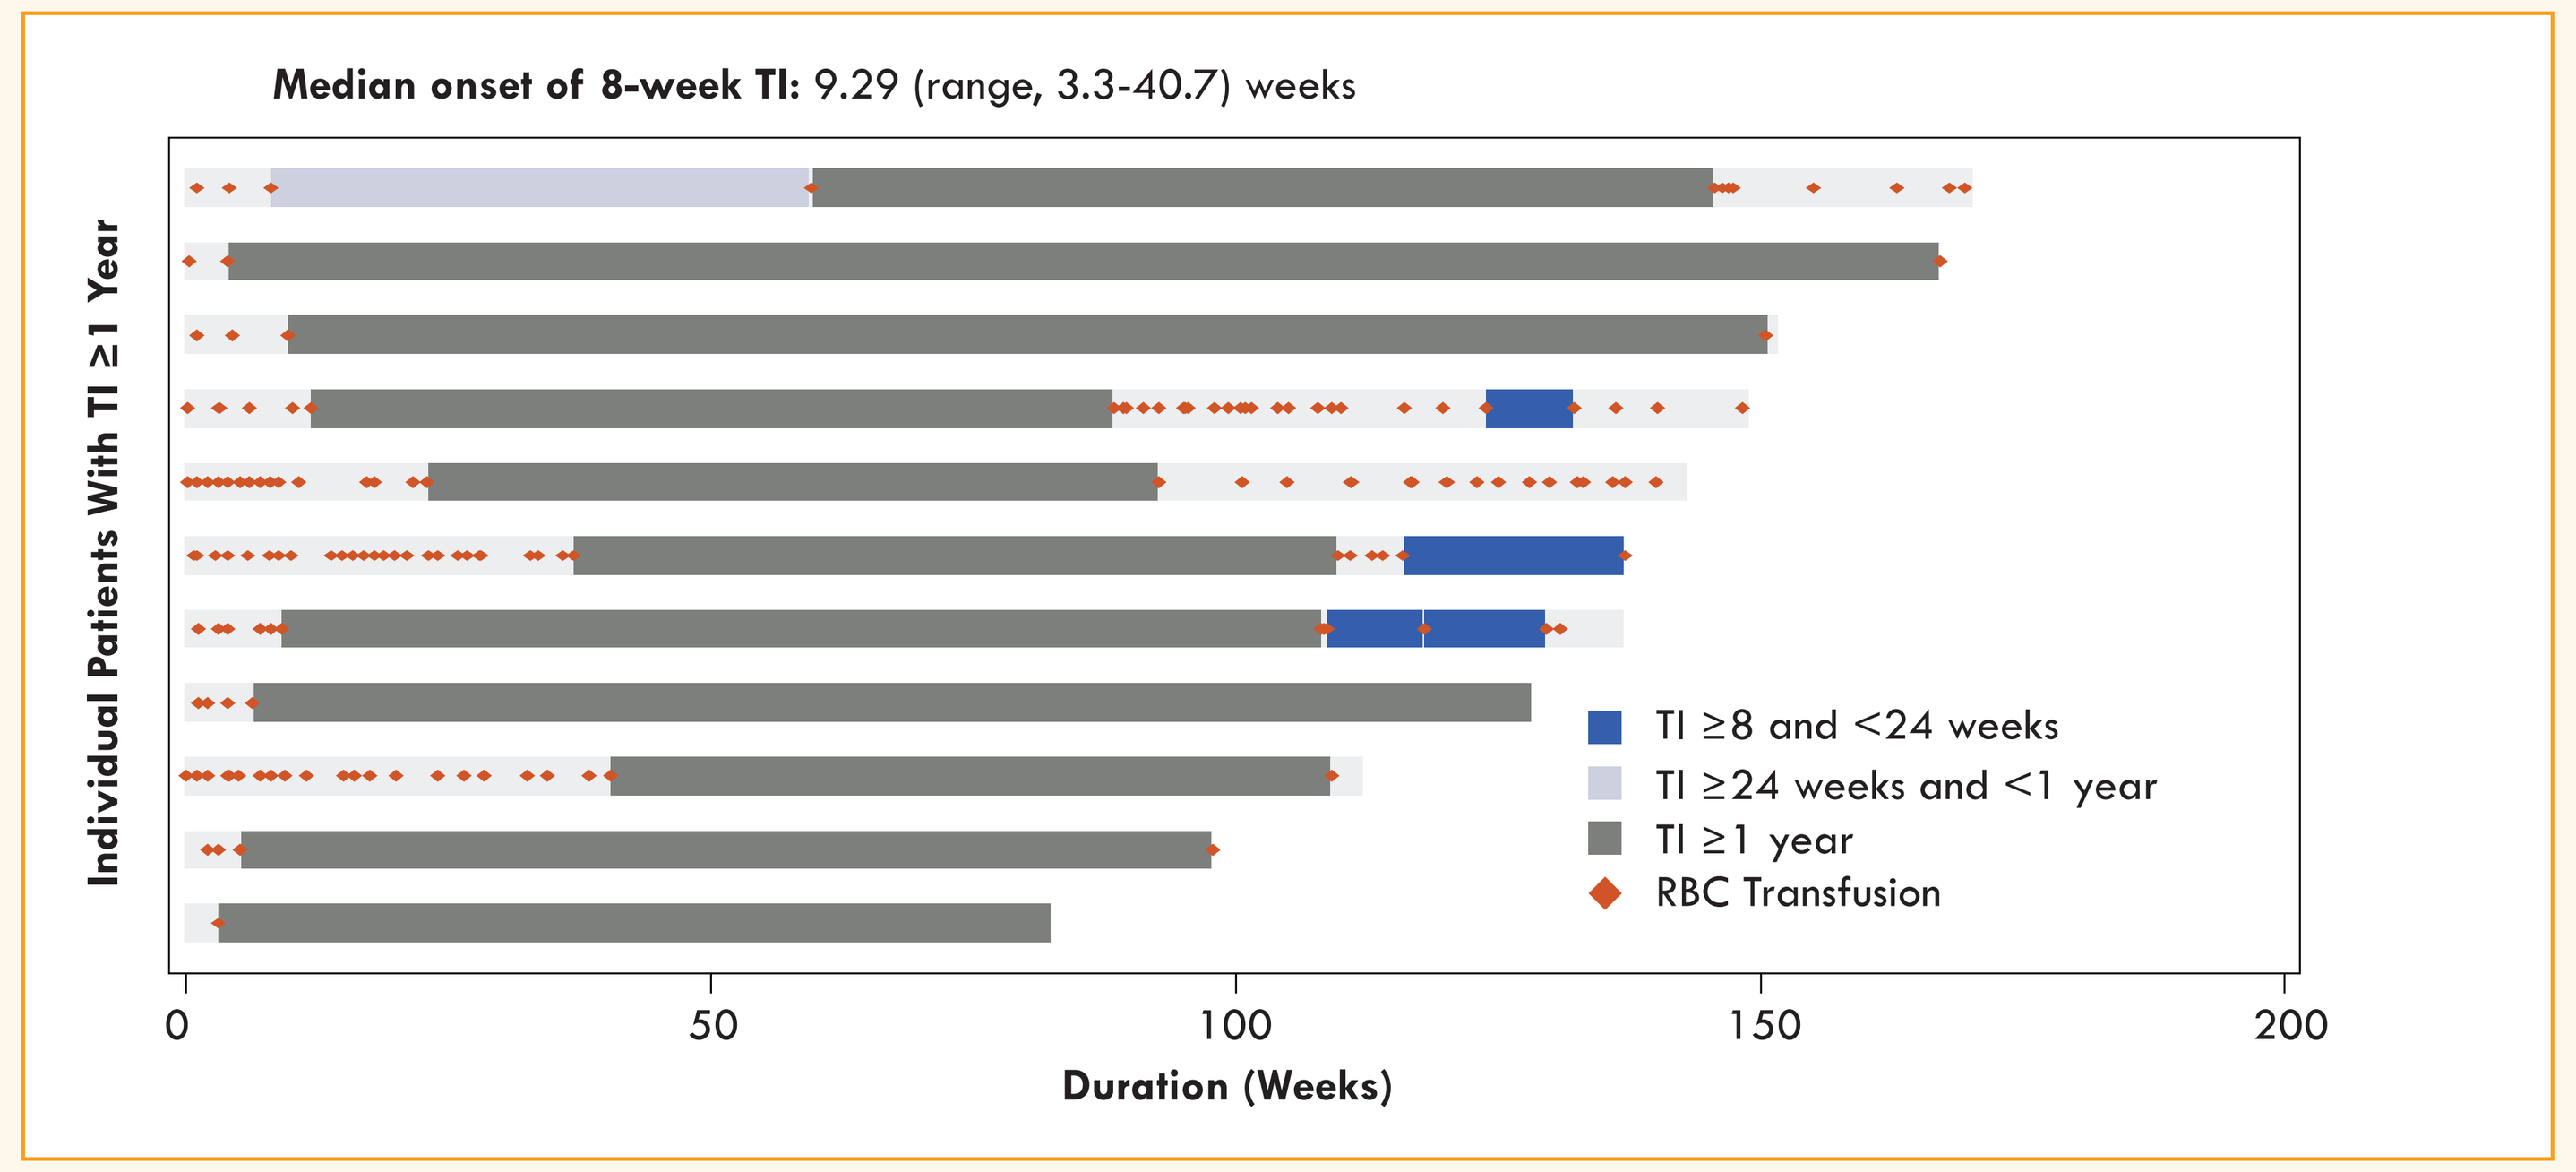
Median onset of 8-week TI: 9.29 (range, 3.3-40.7) weeks
Individual Patients With TI ≥1 Year
TI ≥8 and <24 weeks
TI ≥24 weeks and <1 year
TI ≥1 year
RBC Transfusion
0
50
100
150
200
Duration (Weeks)

LR, lower-risk; MDS, myelodysplastic syndromes; RBC, red blood cell; TI, transfusion independence.

Fig. 4. Durable TI Accompanied by Substantial Increase in Hgb in TI ≥1-Year Responders

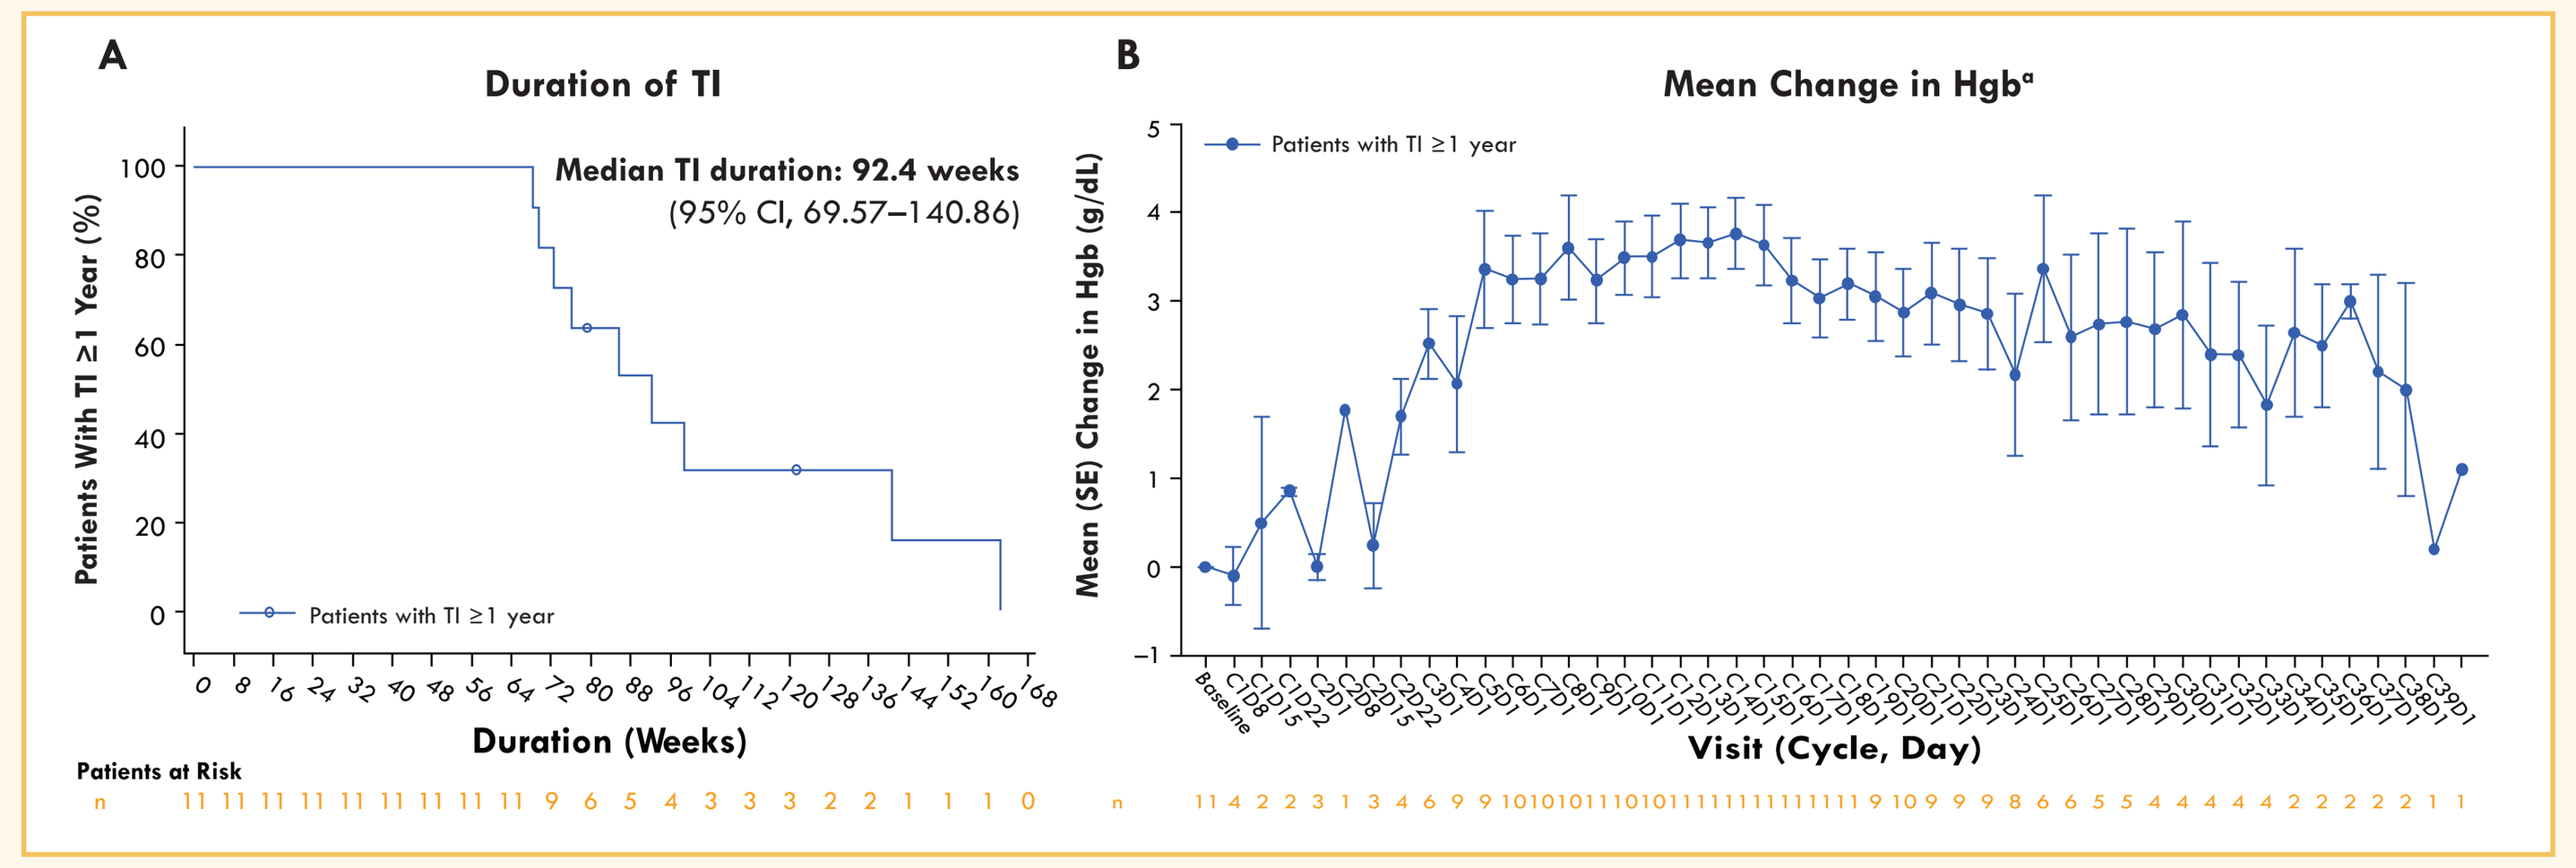
A
Duration of TI
Median TI duration: 92.4 weeks
(95% CI, 69.57–140.86)
Patients With TI ≥1 Year (%)
Patients with TI ≥1 year
Duration (Weeks)
Patients at Risk
n 11 11 11 11 11 11 11 11 11 11 9 6 5 4 3 3 3 2 2 1 1 1 0
B
Mean Change in Hgb[a]
Patients with TI ≥1 year
Mean (SE) Change in Hgb (g/dL)
Visit (Cycle, Day)

[a]The mean changes from the minimum Hgb for values in the 8 weeks prior to the first dose date are shown and values within 14 days of RBC transfusions were excluded. This plot does not include the values from unscheduled visits. Hgb, hemoglobin; RBC, red blood cell; SE, standard error of the mean; TI, transfusion independence.

Mutation data

- Of the 11 TI ≥1-year responders, 9 had pre- and post-treatment mutation data available
 - 8 (89%) demonstrated a reduction in *SF3B1* variant allele frequency (VAF)
 - 5 (56%) achieved ≥50% VAF reduction
- Reduction in VAF correlated with a longer TI duration (median, >20 months and shorter time to TI onset (median, <10 weeks (**Fig. 6**)

Safety

- Overall, safety was consistent between the TI ≥1-year group and the target patient population
- The most frequent adverse events were reversible thrombocytopenia and neutropenia (**Table 3**)
 - >97% of grade 3/4 thrombocytopenia and neutropenia within the ≥1-year TI population resolved to grade 2 or lower within 4 weeks (consistent with target population)
 - Four of the patients with TI ≥1-year had no grade 3 or higher neutropenia/thrombocytopenia
- Events were manageable with dose holds (n=10/11) and reductions (n=7/11) as specified in the protocol with limited clinical consequences
- Imetelstat-related cytopenias are on-target effects based on the selective reduction of malignant cells through telomerase inhibition[8]

Fig. 5. Robust PFS and Survival of Imetelstat-Treated Patients With TI ≥1 Year

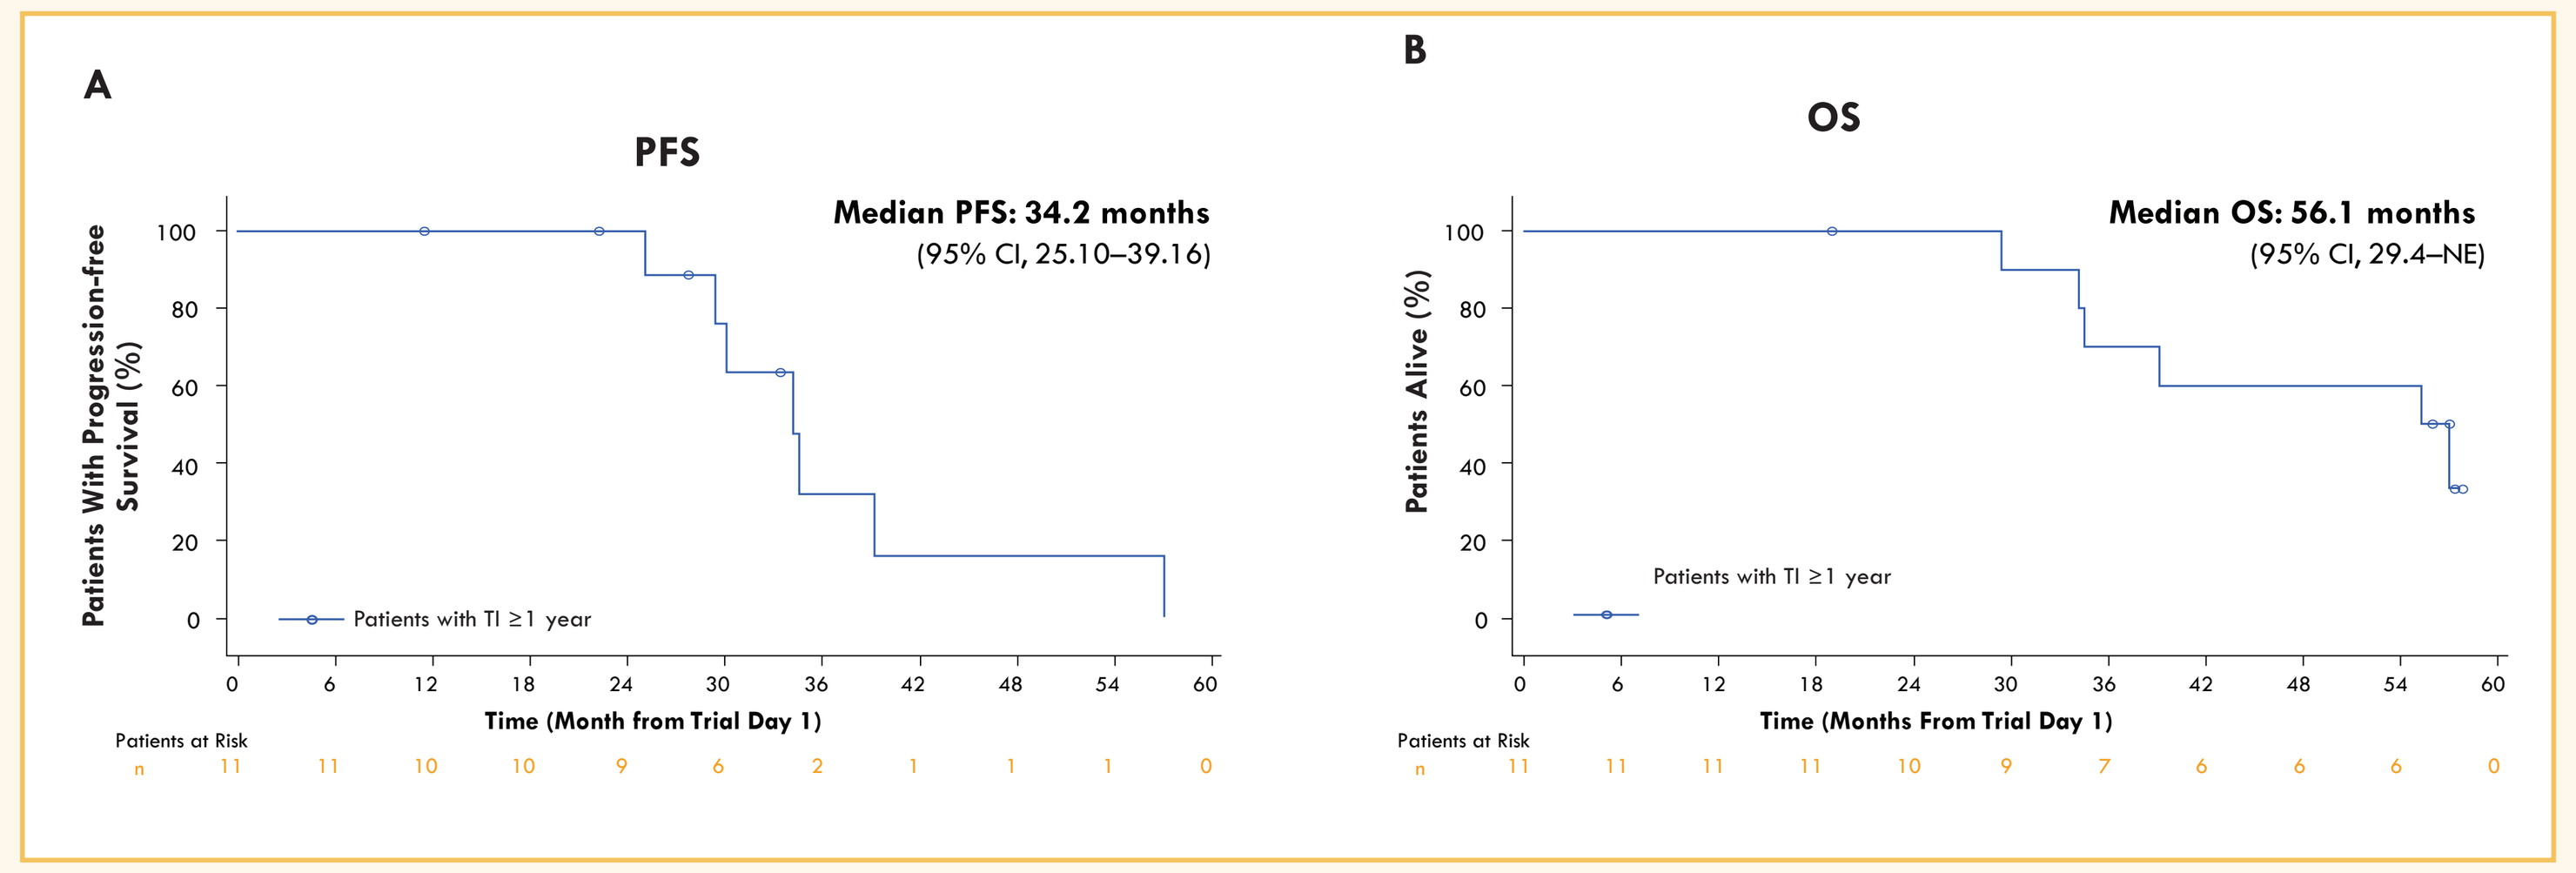
A
PFS
Median PFS: 34.2 months
(95% CI, 25.10–39.16)
Patients With Progression-free Survival (%)
Patients with TI ≥1 year
Time (Month from Trial Day 1)
Patients at Risk
n 11 11 10 10 9 6 2 1 1 1 0
B
OS
Median OS: 56.1 months
(95% CI, 29.4–NE)
Patients Alive (%)
Patients with TI ≥1 year
Time (Months From Trial Day 1)
Patients at Risk
n 11 11 11 11 10 9 7 6 6 6 0

NE, not evaluable; OS, overall survival; PFS, progression-free survival; TI, transfusion independence.

Table 2. Median PFS and OS in TI ≥1 Year Responders and Others in the Target Population

Parameter	Patients with TI ≥1 year (n=11)	Others (n=27)	Target population (N=38)
Median PFS, months (95% CI)	34.2 (25.1, 39.2)	25.5 (11.5, 44.2)	34.2 (25.1, 41.4)
Median OS, months (95% CI)	56.1 (29.4, NE)	47.1 (38.1, NE)	55.2 (38.1, 67.1)

NE, not evaluable; OS, overall survival; PFS, progression-free survival.

Fig. 6. Reduction in *SF3B1* VAF in Imetelstat-Treated Patients With TI ≥1 Year Correlated With Longer TI Duration

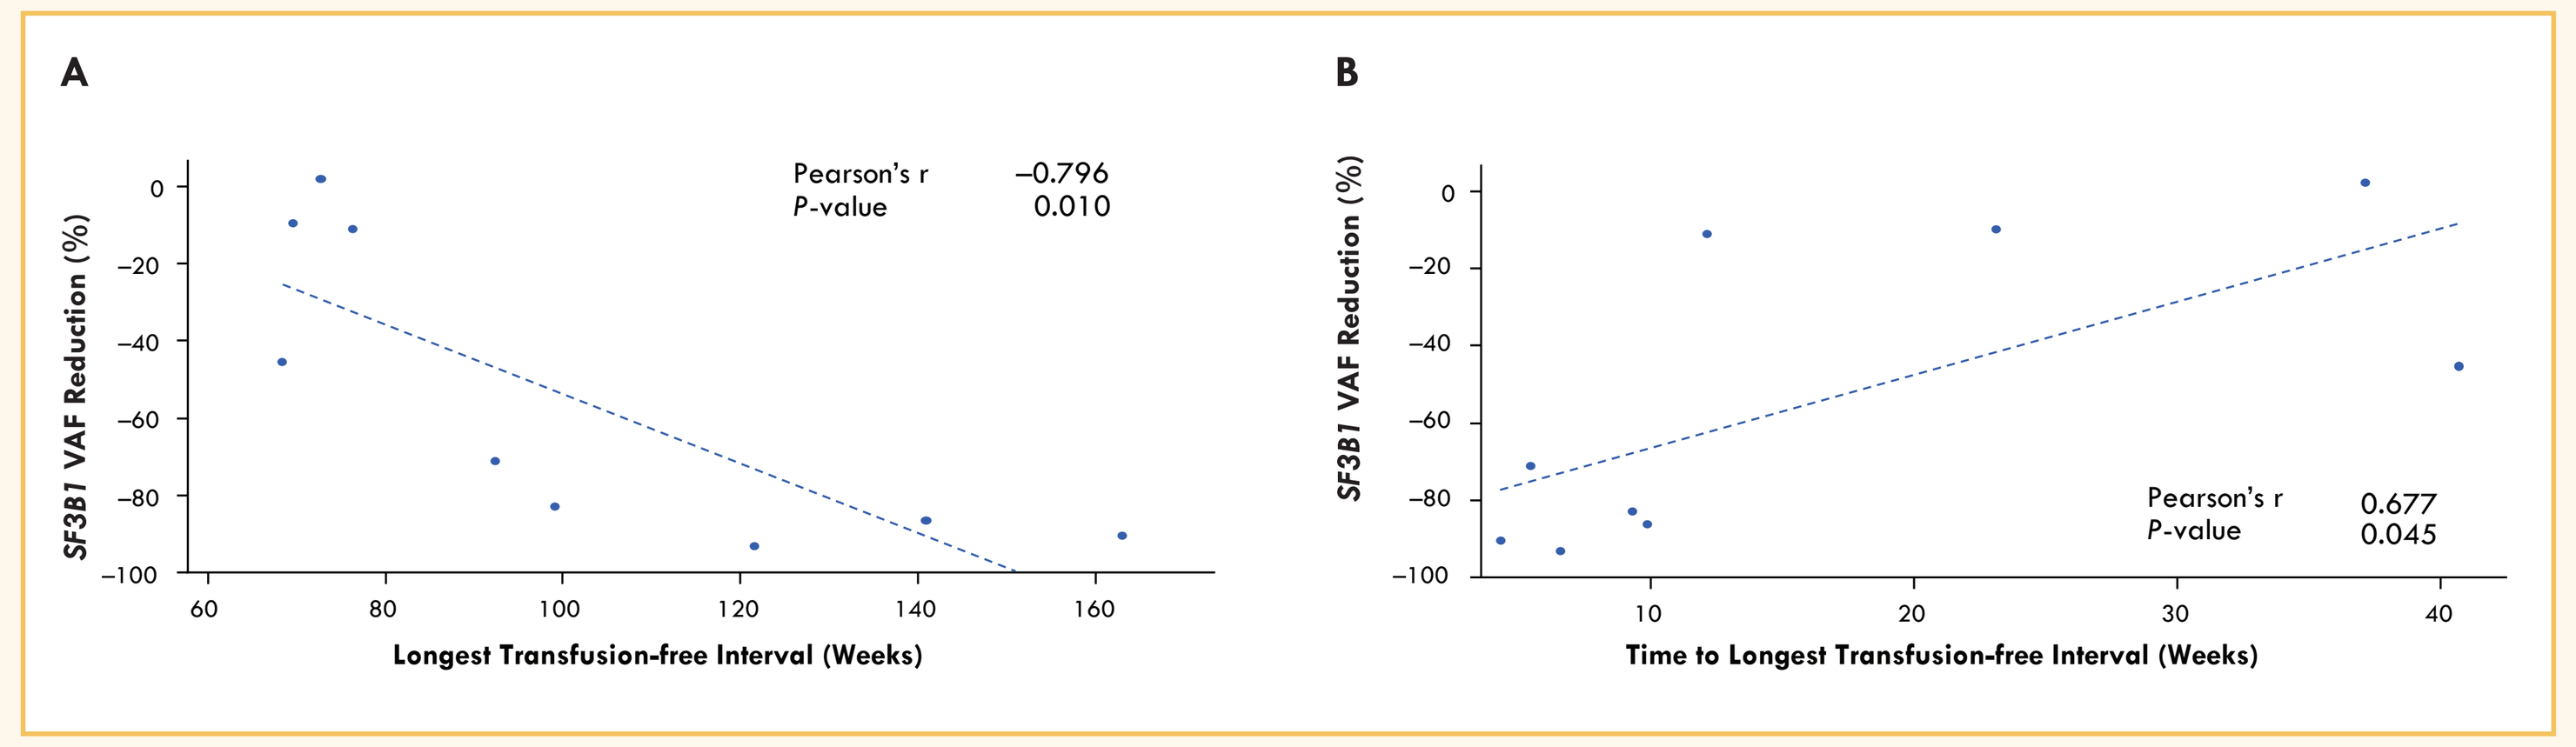
A
Pearson's r −0.796
P-value 0.010
SF3B1 VAF Reduction (%)
Longest Transfusion-free Interval (Weeks)
B
Pearson's r 0.677
P-value 0.045
SF3B1 VAF Reduction (%)
Time to Longest Transfusion-free Interval (Weeks)

TI, transfusion independence; VAF, variant allele frequency.

Table 3. Updated Phase 2 Grade 3/4 Cytopenias Profile in Patients Treated With Imetelstat Similar to Previous Reports[5,8]

Grade 3/4 Cytopenias, n (%)	≥1 year TI n=11	Target population N=38
Thrombocytopenia	7 (63.6)	23 (60.5)
Neutropenia	6 (54.5)	21 (55.3)
Anemia	2 (18.2)	8 (21.2)
Leukopenia	2 (18.2)	7 (18.4)

TI, transfusion independence.

Trial Design

- IMerge (NCT02598661) is a multicenter, global, phase 2/3 trial of imetelstat administered as a 2-hour intravenous (IV) infusion at 7.5 mg/kg every 4 weeks (q4w) in RBC TD, ESA-R/R (or >500 U/L serum erythropoietin) LR-MDS[5,9]
 - Patients were also International Prognostic Scoring System (IPSS) low or intermediate-1, and had ≥4 units RBC/8 weeks over the 16-week pretrial period
- After an initial analysis, the phase 2 trial was expanded to include patients with no prior HMA or len treatments, and a non-del(5q) MDS subtype

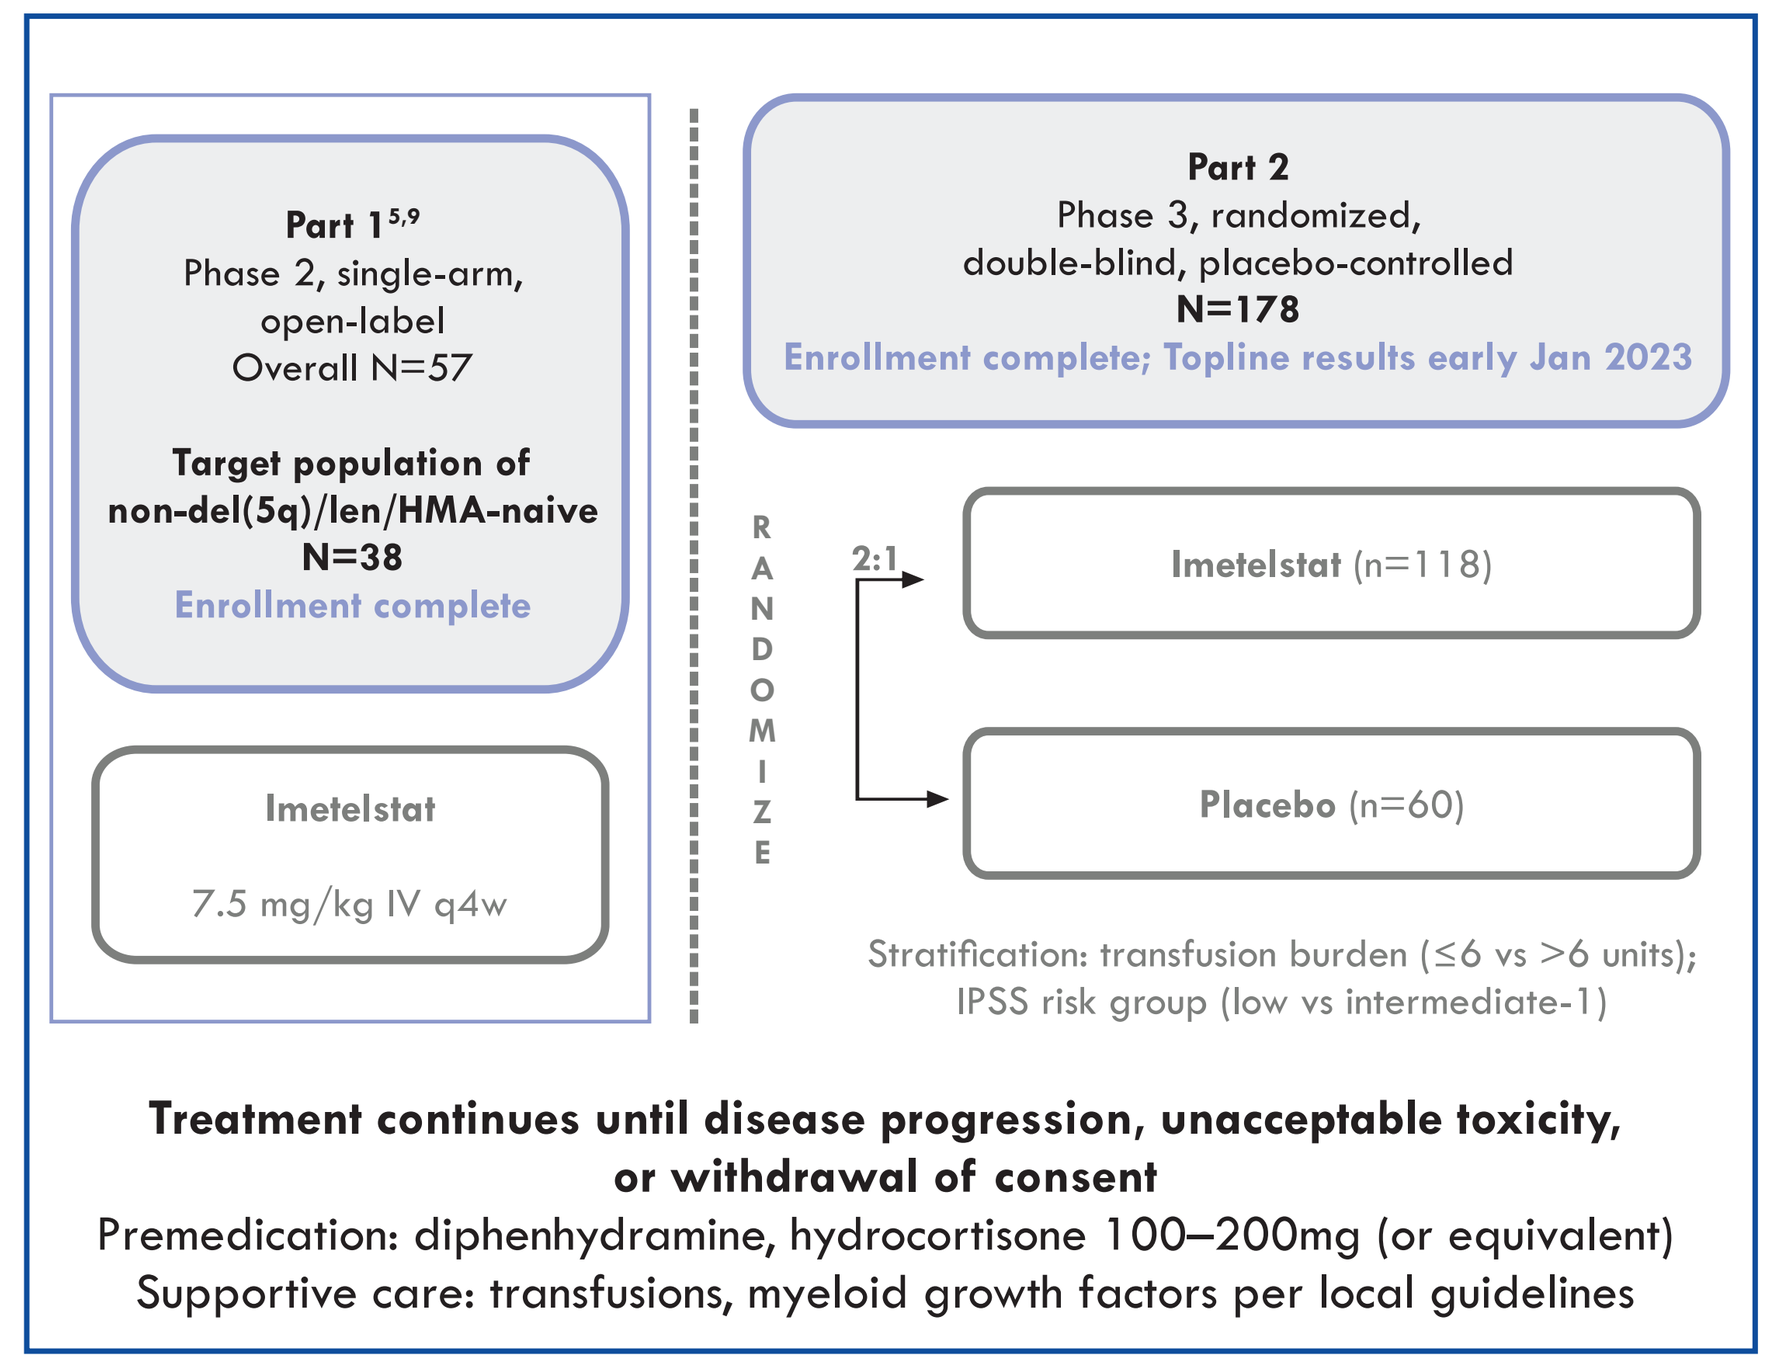
Part 1[5,9]
Phase 2, single-arm,
open-label
Overall N=57
Target population of
non-del(5q)/len/HMA-naive
N=38
Enrollment complete
Imetelstat
7.5 mg/kg IV q4w
Part 2
Phase 3, randomized,
double-blind, placebo-controlled
N=178
Enrollment complete; Topline results early Jan 2023
RANDOMIZE
2:1
Imetelstat (n=118)
Placebo (n=60)
Stratification: transfusion burden (≤6 vs >6 units);
IPSS risk group (low vs intermediate-1)
Treatment continues until disease progression, unacceptable toxicity,
or withdrawal of consent
Premedication: diphenhydramine, hydrocortisone 100–200mg (or equivalent)
Supportive care: transfusions, myeloid growth factors per local guidelines

- The primary endpoint was ≥8-week RBC TI
- Key secondary endpoints included safety, ≥24-week TI rate, hematologic improvement-erythroid (HI-E), overall survival (OS), progression-free survival (PFS), and progression time to AML
 - As per International Working Group criteria, disease progression was defined as at least one of the following: ≥50% decrement from maximum response levels in granulocytes or platelets; ≥1.5 g/dL reduction in hemoglobin (Hgb); transfusion dependence
- The proportion of patients with 8-week, 24-week, and 1-year TI, and other binary endpoints, were summarized with percentage and a 95% 2-sided exact Clopper-Pearson confidence interval (CI)
- The Kaplan-Meier (KM) method was used to estimate the distribution of TI duration, OS, PFS, and progression time to AML

Conclusions

- Imetelstat demonstrated ≥1 year sustained, continuous TI in 29% of patients with transfusion dependent, non-del(5q) LR-MDS relapsed/refractory to ESAs and len/HMA naive
 - Attainment of 24-week TI was indicative of the likelihood to achieve TI ≥1 year
- Strong evidence of disease-modifying activity for imetelstat mechanism of action:
 - Durable TI with median duration of TI of 92.4 weeks and robust increase in Hgb by ≥3 g/dL
 - Notable survival post-ESA (median OS, 56 months)
 - Meaningful reduction in mutational burden that correlated with longer TI and shorter time to onset of TI
- Safety findings were consistent with those of the overall target population and previous reports

ACKNOWLEDGMENTS

- The authors thank all the patients and families for their participation in this trial and acknowledge the collaboration and commitment of all investigators and their staff
 - D. Mazure, S. Meers, D. Breems, M.-P. Gourin, E. Gyan, L. Legros, S. Thepot, I. Kim, J.-H. Lee, A. Pristupa, O. Samoilova, D. Udovitsa, S. Klein, S. Langemeijer, A. van de Loosdrecht, R. De Paz, J. Esteve, D. Valcarcel, B. Xicoy, E. Oliva, R. Boccia, H. Erba, A. DiStast, M. Grunwald, M. Jacoby, C. Miller, G. Schiller, L. Silverman, D. Stevens
- This trial is sponsored by the Geron Corporation
- All authors contributed to and approved the presentation; writing and editorial assistance were provided by Samantha L. Dwyer, PhD, at Ashfield MedComms, an Inizio Company (NJ, USA), funded by the Geron Corporation

DISCLOSURES

U.W.: Honoraria from Geron, Bristol Myers Squibb (BMS)/Celgene, AbbVie, Jazz, Janssen, Silence Therapeutics, and Takeda. R.K.: Speaker bureau from Jazz, Servier, CTI, PharmaEssentia; advisory board fees/honoraria from BMS, Novartis, AbbVie, Jazz, Servier, PharmaEssentia, Taiho, Geron, and CTI. P.F.: Consultancy fees, research funding, and honoraria from BMS, AbbVie, Janssen, Jazz, and Novartis. M.S.: Advisory board fees from Geron, BMS, Novartis, Kurome. M.S: Consultancy fee from Geron. Y.F.M. received honoraria/consulting fees from Blueprint Medicines, Geron, OncLive. YFM participated in advisory boards and received honoraria from Sierra Oncology, Stemline Therapeutics, Blueprint Medicines, Morphosys, Taiho Oncology and Novartis. Y.F.M. received travel reimbursement from Blueprint Medicines and Morphosys. None of these relationships were related to this work. K.V.E: None. A.R.: Consultancy from Taiho and Epizyme. Speaker fees from Celgene, Novartis, Janssen; Research funding from Celgene and Novartis; Advisory fees from Celgene. A.Z. is a Leukemia and Lymphoma Society Scholar in Clinical Research. A.Z. received research funding (institutional) from Celgene/BMS, AbbVie, Astex, Pfizer, Medimmune/AstraZeneca, Boehringer Ingelheim, Cardiff Oncology, Incyte, Takeda, Novartis, Aprea, and ADC Therapeutics. A.Z. participated in advisory boards, and/or had a consultancy with and received honoraria from AbbVie, Otsuka, Pfizer, Celgene/BMS, Jazz, Incyte, Agios, Boehringer Ingelheim, Novartis, Acceleron, Astellas, Daiichi Sankyo, Cardinal Health, Taiho, Seattle Genetics, BeyondSpring, Cardiff Oncology, Takeda, Ionis, Amgen, Janssen, Epizyme, Syndax, Gilead, Kura, Chiesi, ALX Oncology, BioCryst, Notable, Orum, and Tyme. A.Z. served on clinical trial committees for Novartis, AbbVie, Gilead, BioCryst, AbbVie, ALX Oncology, Geron and Celgene/BMS. A.Z. received travel support for meetings from Pfizer, Novartis, and Cardiff Oncology; consulting/honoraria and research funding from Geron. V.S.: Advisory board: Abbvie, Geron, Gilead, BMS, Menarini, Novartis, Servier, and Syros. Travel grant: Janssen. S.S, L.S., A.I., F.F: Current employment and current equity holder in publicly traded company: Geron Corporation.

REFERENCES

1. Asai A, et al. *Cancer Res.* 2003;63(14):3931–3939
2. Herbert BS, et al. *Oncogene.* 2005;24(33):5262–5268
3. Mosoyan G, et al. *Leukemia.* 2017;31(11):2458–2467
4. Wang X at al. *Blood Adv.* 2018;25;2(18):2378–2388
5. Steensma DP, et al. *J Clin Oncol.* 2021;39(1):48–56
6. Platzbecker U, et al. Presented at: ASH Annual Meeting 2020; Abstract 658
7. Geron Announces Positive Top-Line Results from IMerge Phase 3 Trial of Imetelstat in Lower Risk MDS.; 2023. Available at: www.ir.geron.com. Accessed January 4, 2023
8. Mascarenhas J, et al. Presented at: EHA Annual Meeting 2021; Abstract EP0116
9. Platzbecker U, et al. Presented at: ASH Annual Meeting 2020; Abstract 3113

Originally presented as an oral presentation at the American Society of Hematology Annual Meeting, December 10–13, 2022, in New Orleans, LA, United States. Adapted with permission and presented at the 27th Annual International Congress on Hematologic Malignancies®: Focus on Leukemias, Lymphomas, and Myeloma, February 23–26, 2023, in Miami Beach, FL, United States.